Deucravacitinib long-term efficacy with continuous treatment in plaque psoriasis: 2-year results from the phase 3 POETYK PSO-1 and PSO-2 trials

Diamant Thaçi,[1] Mark Lebwohl,[2] Richard B. Warren,[3] Leon Kircik,[2] Joannee Zumkehr,[4] Subhashis Banerjee,[4] Andrew Napoli,[4] Renata M. Kisa,[4] Misti Linaberry,[4] Kim Hoyt,[4] Andrew Blauvelt[5]

[1]Institute and Comprehensive Center for Inflammation Medicine, University of Lübeck, Lübeck, Germany; [2]Icahn School of Medicine at Mount Sinai, New York, NY, USA; [3]Dermatology Centre, Northern Care Alliance NHS Foundation Trust, NIHR Manchester Biomedical Research Centre, Manchester University NHS Foundation Trust, Manchester Academic Health Science Centre, Manchester, UK; [4]Bristol Myers Squibb, Princeton, NJ, USA; [5]Oregon Medical Research Center, Portland, OR, USA

Background

- Tyrosine kinase 2 (TYK2) is an intracellular enzyme that mediates signaling of cytokines (eg, interleukin-23, Type I interferons) that are involved in psoriasis pathogenesis[1]
- Deucravacitinib, an oral, selective, allosteric TYK2 inhibitor, is approved in the US, EU, and other countries for the treatment of adults with moderate-to-severe plaque psoriasis who are candidates for systemic therapy[2-6]
- Deucravacitinib uniquely binds to the regulatory domain of TYK2 rather than to the catalytic domain where Janus kinase (JAK) 1,2,3 inhibitors bind[1,7] (**Figure 1**), representing the first in a new class of small molecules

Figure 1. Mechanism of action of deucravacitinib

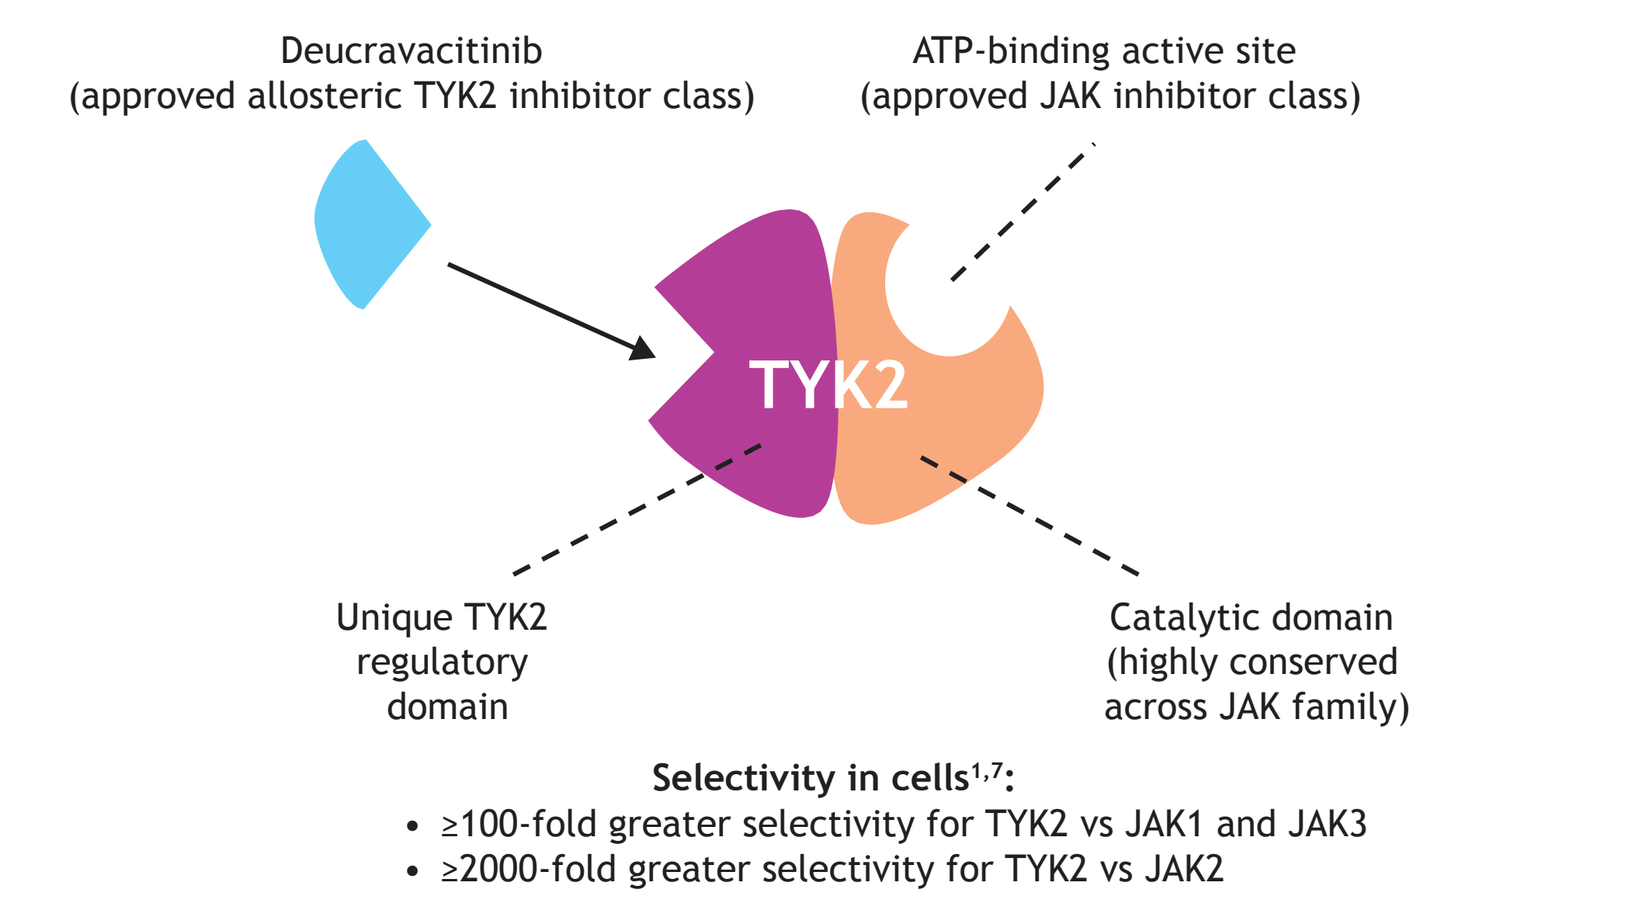

Deucravacitinib
(approved allosteric TYK2 inhibitor class)
ATP-binding active site
(approved JAK inhibitor class)
TYK2
Unique TYK2 regulatory domain
Catalytic domain (highly conserved across JAK family)
Selectivity in cells[1,7]:
• ≥100-fold greater selectivity for TYK2 vs JAK1 and JAK3
• ≥2000-fold greater selectivity for TYK2 vs JAK2

ATP, 5′-adenosine triphosphate; JAK, Janus kinase; TYK2, tyrosine kinase 2.

- Two phase 3 trials, POETYK PSO-1 (NCT03624127) and POETYK PSO-2 (NCT03611751), demonstrated that deucravacitinib was significantly more efficacious than placebo (based on the coprimary endpoints of ≥75% reduction from baseline in Psoriasis Area and Severity Index [PASI 75] and static Physician Global Assessment score of 0 [clear] or 1 [almost clear] with a ≥2-point improvement from baseline [sPGA 0/1]) and apremilast at Week 16 and was well tolerated in patients with moderate to severe plaque psoriasis[8,9]
 - Clinical responses were maintained through 52 weeks in patients who received continuous deucravacitinib treatment from baseline[10]
 - Patients completing POETYK PSO-1 and PSO-2 could enroll in the POETYK long-term extension (LTE) trial (NCT04036435)

Objective

- To evaluate long-term efficacy responses (up to ≈2 years) in patients who were randomized to deucravacitinib in POETYK PSO-1 or PSO-2 and who entered the POETYK LTE trial:
 - Patients who received continuous deucravacitinib treatment from baseline
 - Patients who received continuous deucravacitinib treatment from baseline and who achieved PASI 75 at Week 24

Methods

Study designs

- POETYK PSO-1 and PSO-2 were 52-week, phase 3, double-blind trials that randomized adults with moderate to severe plaque psoriasis 1:2:1 to oral placebo, deucravacitinib 6 mg once daily, or apremilast 30 mg twice daily (**Figure 2**)
 - Patients were diagnosed with moderate to severe plaque psoriasis based on PASI ≥12, sPGA ≥3, and body surface area (BSA) involvement ≥10%
- At Week 52, patients were eligible to enroll in the POETYK LTE trial and receive open-label deucravacitinib 6 mg once daily

Figure 2. POETYK PSO-1, PSO-2, and LTE study designs

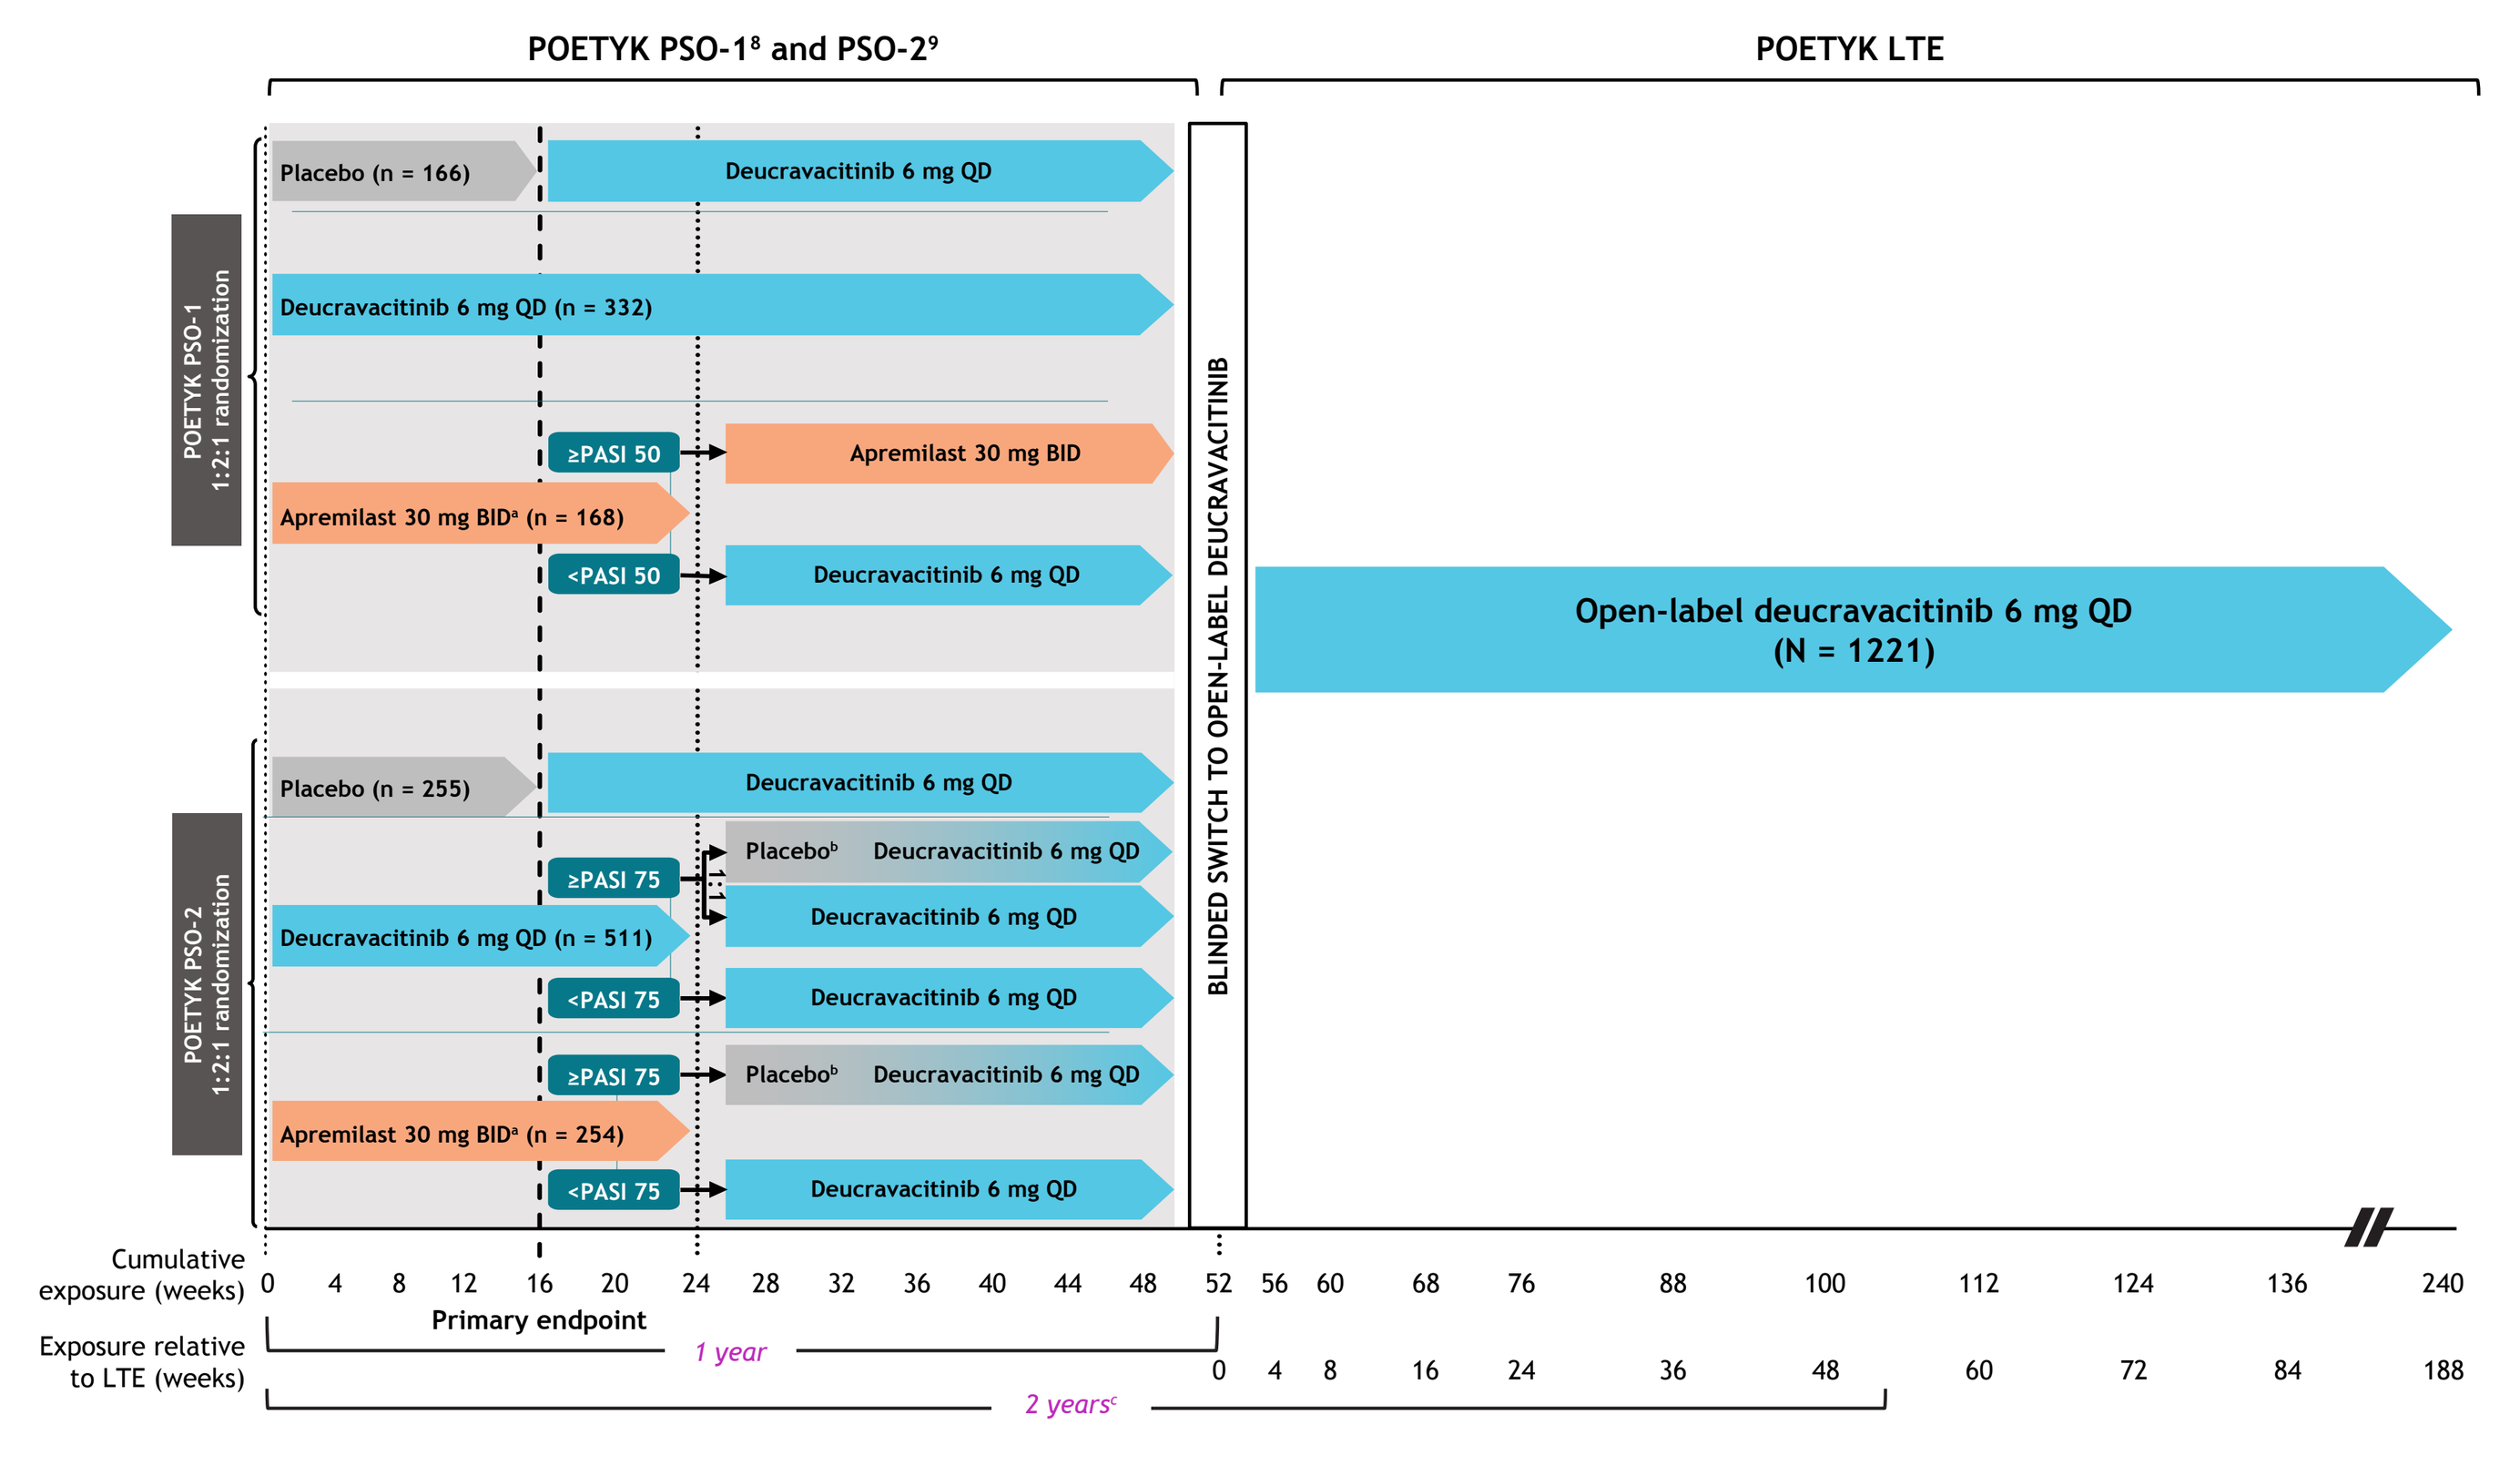

POETYK PSO-1[8] and PSO-2[9]
POETYK LTE
POETYK PSO-1 1:2:1 randomization
Placebo (n = 166)
Deucravacitinib 6 mg QD
Deucravacitinib 6 mg QD (n = 332)
≥PASI 50
Apremilast 30 mg BID
Apremilast 30 mg BID[a] (n = 168)
<PASI 50
Deucravacitinib 6 mg QD
POETYK PSO-2 1:2:1 randomization
Placebo (n = 255)
Deucravacitinib 6 mg QD
≥PASI 75
Placebo[b] Deucravacitinib 6 mg QD
Deucravacitinib 6 mg QD
Deucravacitinib 6 mg QD (n = 511)
<PASI 75
Deucravacitinib 6 mg QD
≥PASI 75
Placebo[b] Deucravacitinib 6 mg QD
Apremilast 30 mg BID[a] (n = 254)
<PASI 75
Deucravacitinib 6 mg QD
BLINDED SWITCH TO OPEN-LABEL DEUCRAVACITINIB
Open-label deucravacitinib 6 mg QD
(N = 1221)
Cumulative exposure (weeks)
0 4 8 12 16 20 24 28 32 36 40 44 48 52 56 60 68 76 88 100 112 124 136 240
Primary endpoint
Exposure relative to LTE (weeks)
0 4 8 16 24 36 48 60 72 84 188
1 year
2 years[c]

[a]Apremilast was titrated from 10 mg QD to 30 mg BID over the first 5 days of dosing. [b]Upon relapse (≥50% loss of Week 24 PASI percent improvement from baseline), patients were to be switched to deucravacitinib 6 mg QD; however, due to a programming error, these patients continued to receive placebo until Week 52. [c]Data reported through the 120-day POETYK LTE trial cutoff date of October 1, 2021.
BID, twice daily; LTE, long-term extension; PASI, Psoriasis Area and Severity Index; PASI 50, ≥50% reduction from baseline in PASI; PASI 75, ≥75% reduction from baseline in PASI; QD, once daily.

Efficacy population

- Pooled POETYK PSO-1 and PSO-2 patient populations
 - Overall analysis: patients who received continuous deucravacitinib treatment from Day 1 until Week 112
 - Responder analysis: Week 24 PASI 75 responders who received continuous deucravacitinib treatment from Day 1 until Week 112
 - Week 24 was chosen as the time point for peak responses observed with deucravacitinib
 - In POETYK PSO-2, only patients who achieved PASI 75 and continued on deucravacitinib at Week 24 (50% of PASI 75 responders based on study design) were analyzed

Efficacy outcomes

- Achievement of PASI 75, ≥90% reduction from baseline in PASI (PASI 90), and sPGA 0/1

Statistical analysis

- Analyses were conducted through the data cutoff date of October 1, 2021 (Week 112)
- Consistent with recommendations for assessing long-term efficacy outcomes in psoriasis clinical trials,[11] 2 methods for imputation of missing data were used as sensitivity analyses in addition to as observed values:
 - **Modified nonresponder imputation (mNRI)[12]:** patients who discontinued treatment due to worsening of psoriasis were imputed as nonresponders; other missing data were analyzed by multiple imputation
 - **Treatment failure rules (TFR)[13]:** patients who discontinued treatment due to lack of efficacy, worsening of psoriasis, or use of a protocol-prohibited medication were imputed as nonresponders

Results

Baseline patient demographics and disease characteristics

- A total of 513 patients received continuous deucravacitinib treatment from Day 1 in POETYK PSO-1/PSO-2 until Week 112 in POETYK LTE (**Table 1**), including 336 patients who had achieved PASI 75 at Week 24 (PSO-1, n = 203; PSO-2, n = 133)
 - Baseline patient demographics and disease characteristics were similar in both groups

Table 1. Baseline patient demographics and disease characteristics

Parameter	Deucravacitinib (n = 513)	Deucravacitinib Week 24 PASI 75 responders (n = 336)
Age, mean (SD), y	46.9 (13.3)	46.3 (13.8)
Weight, mean (SD), kg	89.9 (22.2)	86.6 (22.1)
Female, n (%)	159 (31.0)	122 (36.3)
Race, n (%)		
White	440 (85.8)	284 (84.5)
Asian	64 (12.5)	47 (14.0)
Black or African American	5 (1.0)	1 (0.3)
Other	4 (0.8)	4 (1.2)
Age at disease onset, mean (SD), y	29.0 (14.7)	28.8 (15.3)
Disease duration, mean (SD), y	18.8 (12.6)	18.3 (13.0)
PASI score, mean (SD)	21.1 (7.9)	21.3 (8.0)
sPGA score, n (%)		
3 (moderate)	401 (78.2)	265 (78.9)
4 (severe)	112 (21.8)	71 (21.1)
BSA involvement, mean (SD), %	26.9 (15.8)	27.0 (15.8)

BSA, body surface area; PASI, Psoriasis Area and Severity Index; PASI 75, ≥75% reduction from baseline in PASI; SD, standard deviation; sPGA, static Physician Global Assessment.

Efficacy of continuous deucravacitinib treatment from Day 1

- High clinical response rates achieved at Week 52 in POETYK PSO-1/PSO-2 were maintained at Week 112 in the POETYK LTE trial among patients receiving continuous deucravacitinib treatment from Day 1 (mNRI) (**Figures 3-5**)
 - PASI 75: Week 52, 73.0%; Week 112, 76.2%
 - PASI 90: Week 52, 46.3%; Week 112, 48.3%
 - sPGA 0/1: Week 52, 58.5%; Week 112, 58.4%
- Results were consistent regardless of data imputation methodology (mNRI, TFR, as-observed values)

Figure 3. PASI 75 response rates in patients who received continuous deucravacitinib from baseline until Week 112

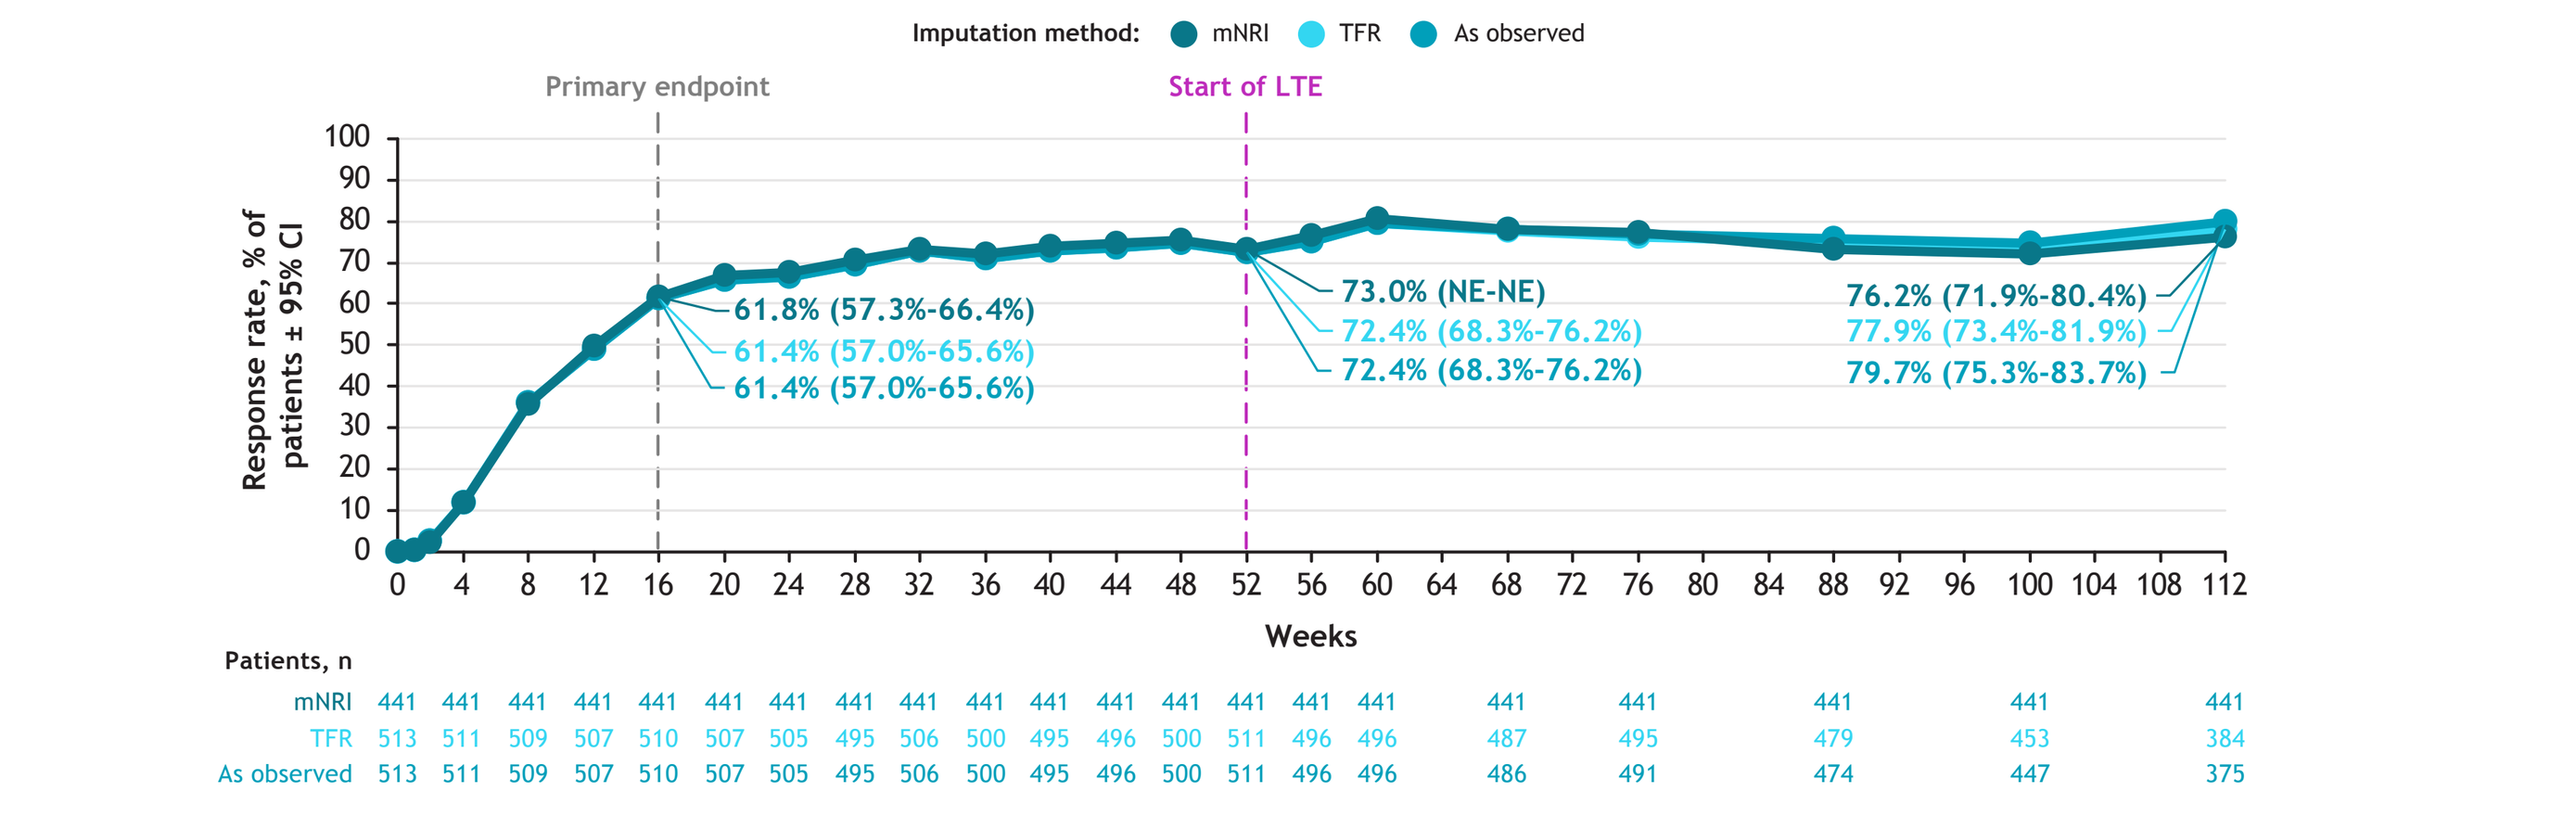

Imputation method: mNRI TFR As observed
Primary endpoint
Start of LTE
Response rate, % of patients ± 95% CI
61.8% (57.3%-66.4%)
61.4% (57.0%-65.6%)
61.4% (57.0%-65.6%)
73.0% (NE-NE)
72.4% (68.3%-76.2%)
72.4% (68.3%-76.2%)
76.2% (71.9%-80.4%)
77.9% (73.4%-81.9%)
79.7% (75.3%-83.7%)
Weeks
Patients, n
mNRI 441 441 441 441 441 441 441 441 441 441 441 441 441 441 441 441 441 441 441 441 441
TFR 513 511 509 507 510 507 505 495 506 500 495 496 500 511 496 496 487 495 479 453 384
As observed 513 511 509 507 510 507 505 495 506 500 495 496 500 511 496 496 486 491 474 447 375

CI, confidence interval; LTE, long-term extension; mNRI, modified nonresponder imputation; NE, not estimable; PASI 75, ≥75% reduction from baseline in Psoriasis Area and Severity Index; TFR, treatment failure rules.

Figure 4. PASI 90 response rates in patients who received continuous deucravacitinib treatment from baseline until Week 112

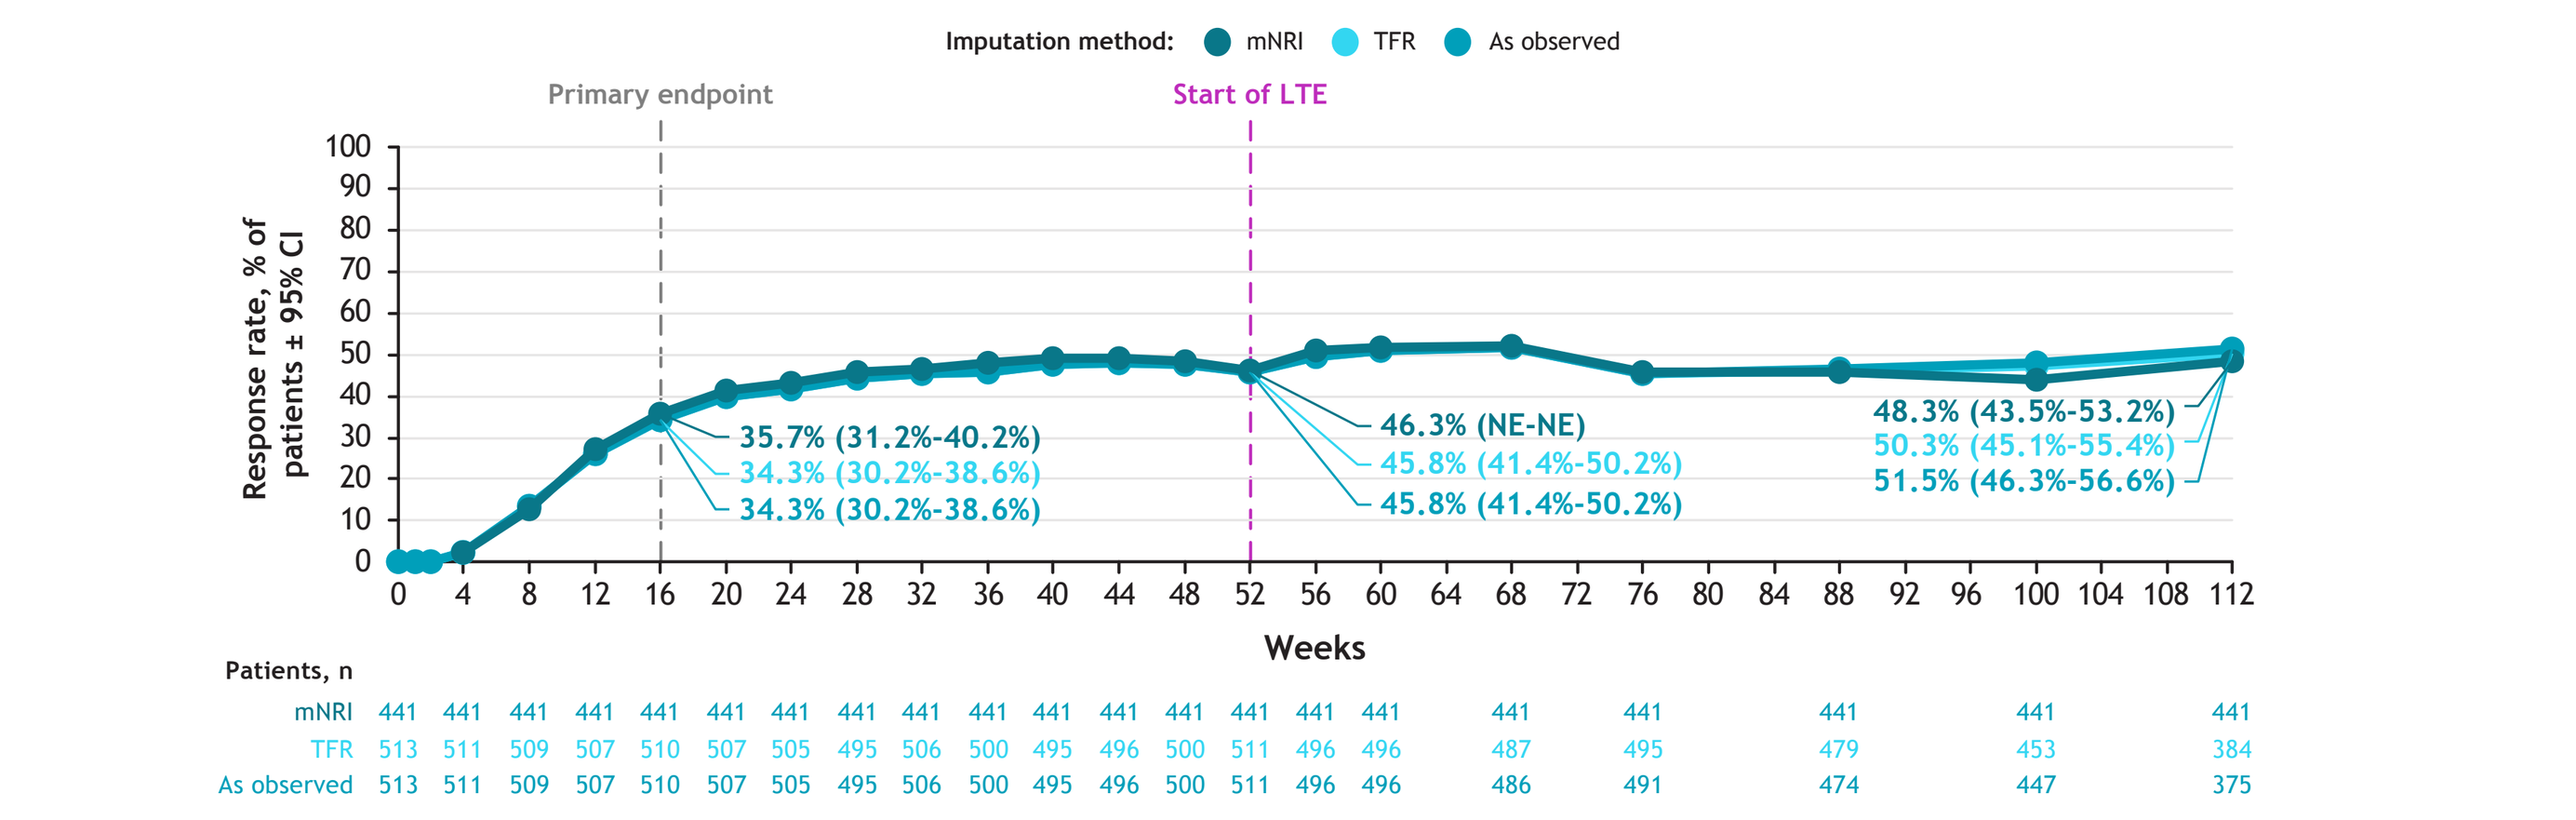

Imputation method: mNRI TFR As observed
Primary endpoint
Start of LTE
Response rate, % of patients ± 95% CI
35.7% (31.2%-40.2%)
34.3% (30.2%-38.6%)
34.3% (30.2%-38.6%)
46.3% (NE-NE)
45.8% (41.4%-50.2%)
45.8% (41.4%-50.2%)
48.3% (43.5%-53.2%)
50.3% (45.1%-55.4%)
51.5% (46.3%-56.6%)
Weeks
Patients, n
mNRI 441 441 441 441 441 441 441 441 441 441 441 441 441 441 441 441 441 441 441 441 441
TFR 513 511 509 507 510 507 505 495 506 500 495 496 500 511 496 496 487 495 479 453 384
As observed 513 511 509 507 510 507 505 495 506 500 495 496 500 511 496 496 486 491 474 447 375

CI, confidence interval; LTE, long-term extension; mNRI, modified nonresponder imputation; NE, not estimable; PASI 90, ≥90% reduction from baseline in Psoriasis Area and Severity Index; TFR, treatment failure rules.

Figure 5. sPGA 0/1 response rates in patients who received continuous deucravacitinib treatment from baseline until Week 112

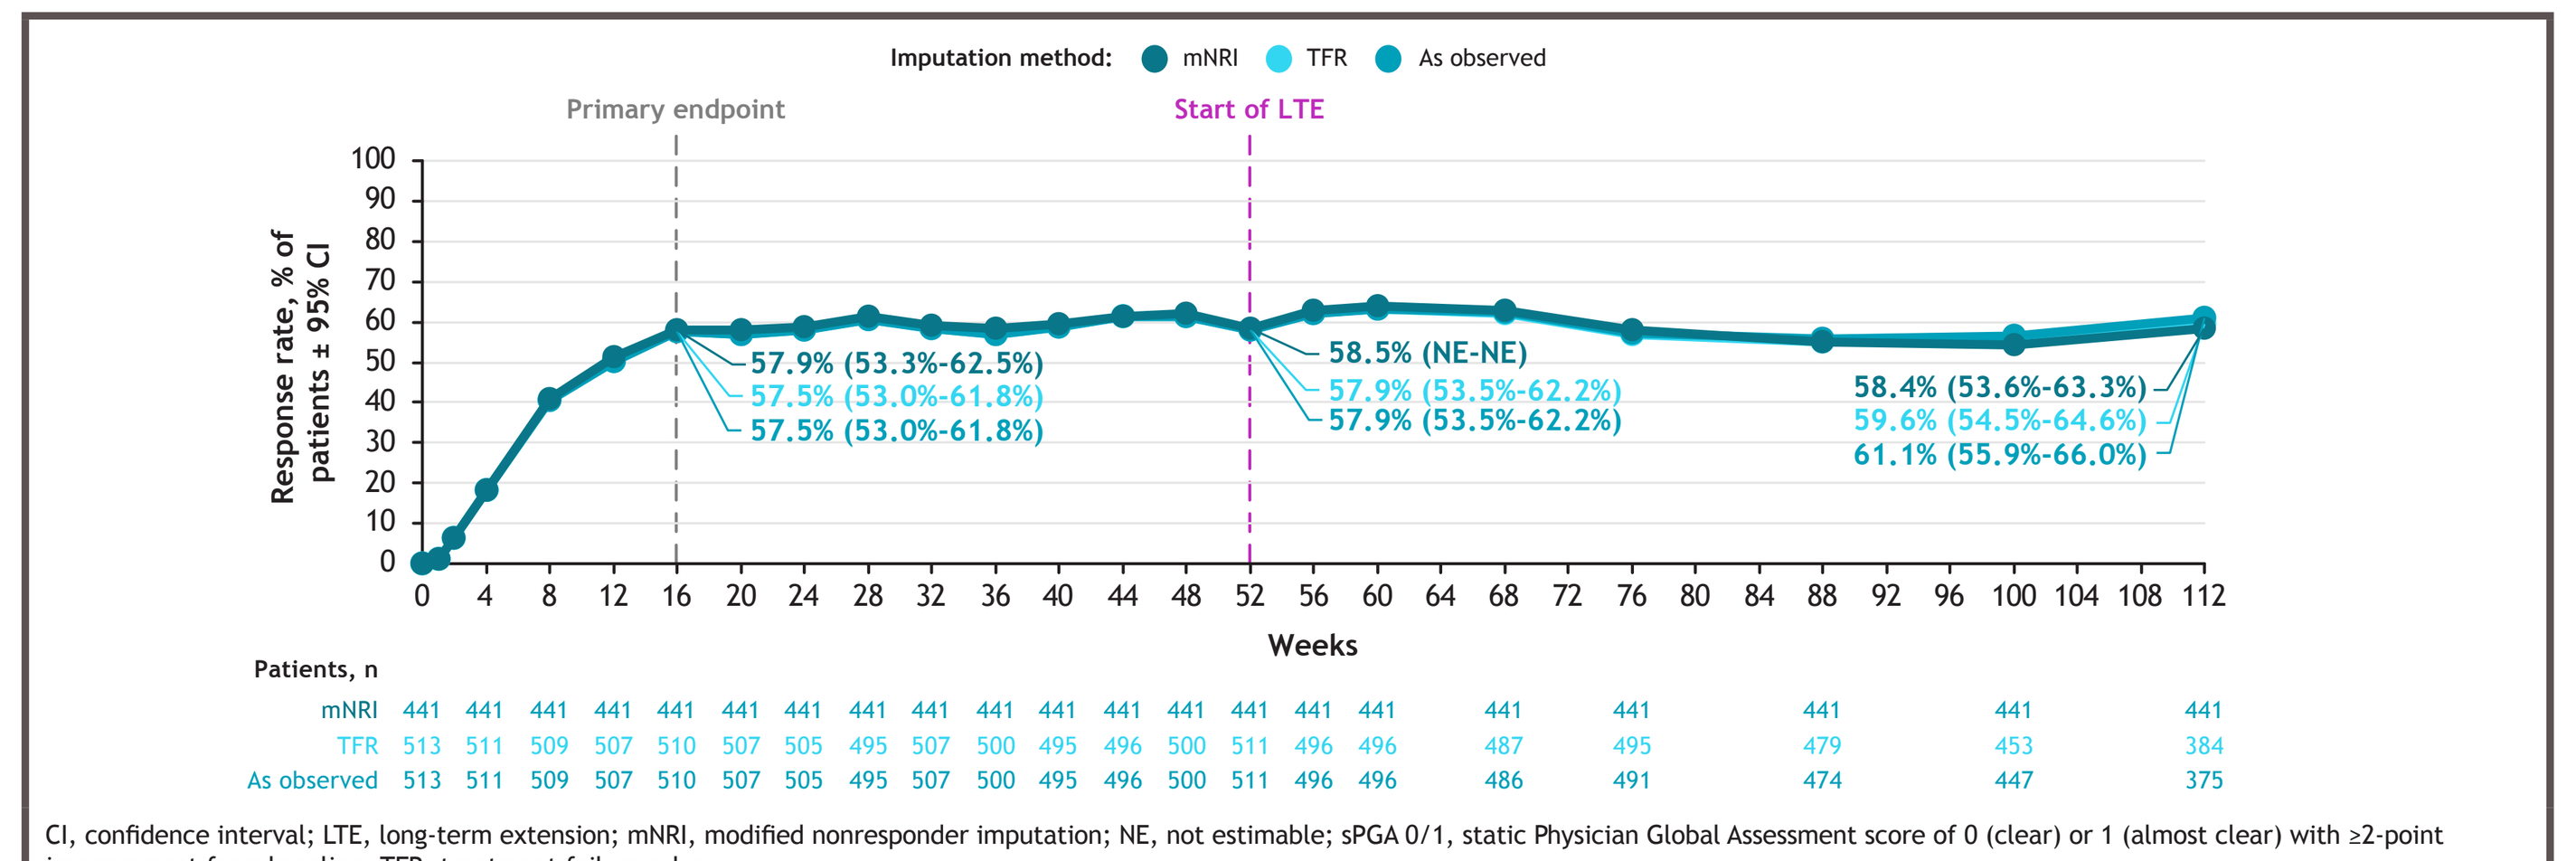

Imputation method: mNRI TFR As observed
Primary endpoint
Start of LTE
Response rate, % of patients ± 95% CI
57.9% (53.3%-62.5%)
57.5% (53.0%-61.8%)
57.5% (53.0%-61.8%)
58.5% (NE-NE)
57.9% (53.5%-62.2%)
57.9% (53.5%-62.2%)
58.4% (53.6%-63.3%)
59.6% (54.5%-64.6%)
61.1% (55.9%-66.0%)
Weeks
Patients, n
mNRI 441 441 441 441 441 441 441 441 441 441 441 441 441 441 441 441 441 441 441 441 441
TFR 513 511 509 507 510 507 505 495 507 500 495 496 500 511 496 496 487 495 479 453 384
As observed 513 511 509 507 510 507 505 495 507 500 495 496 500 511 496 496 486 491 474 447 375

CI, confidence interval; LTE, long-term extension; mNRI, modified nonresponder imputation; NE, not estimable; sPGA 0/1, static Physician Global Assessment score of 0 (clear) or 1 (almost clear) with ≥2-point improvement from baseline; TFR, treatment failure rules.

Efficacy in Week 24 PASI 75 responders

- High response rates at Week 52 in POETYK PSO-1/PSO-2 were maintained at Week 112 in the POETYK LTE among patients receiving continuous deucravacitinib treatment from Day 1 who had achieved PASI 75 at Week 24 (mNRI) (**Figures 6-8**)
 - PASI 75: Week 52, 90.3%; Week 112, 89.2%
 - PASI 90: Week 52, 62.1%; Week 112, 61.1%
 - sPGA 0/1: Week 52, 73.8%; Week 112, 70.5%
- Results were consistent regardless of data imputation methodology

Figure 6. PASI 75 response rates in Week 24 PASI 75 responders who received continuous deucravacitinib treatment from baseline until Week 112

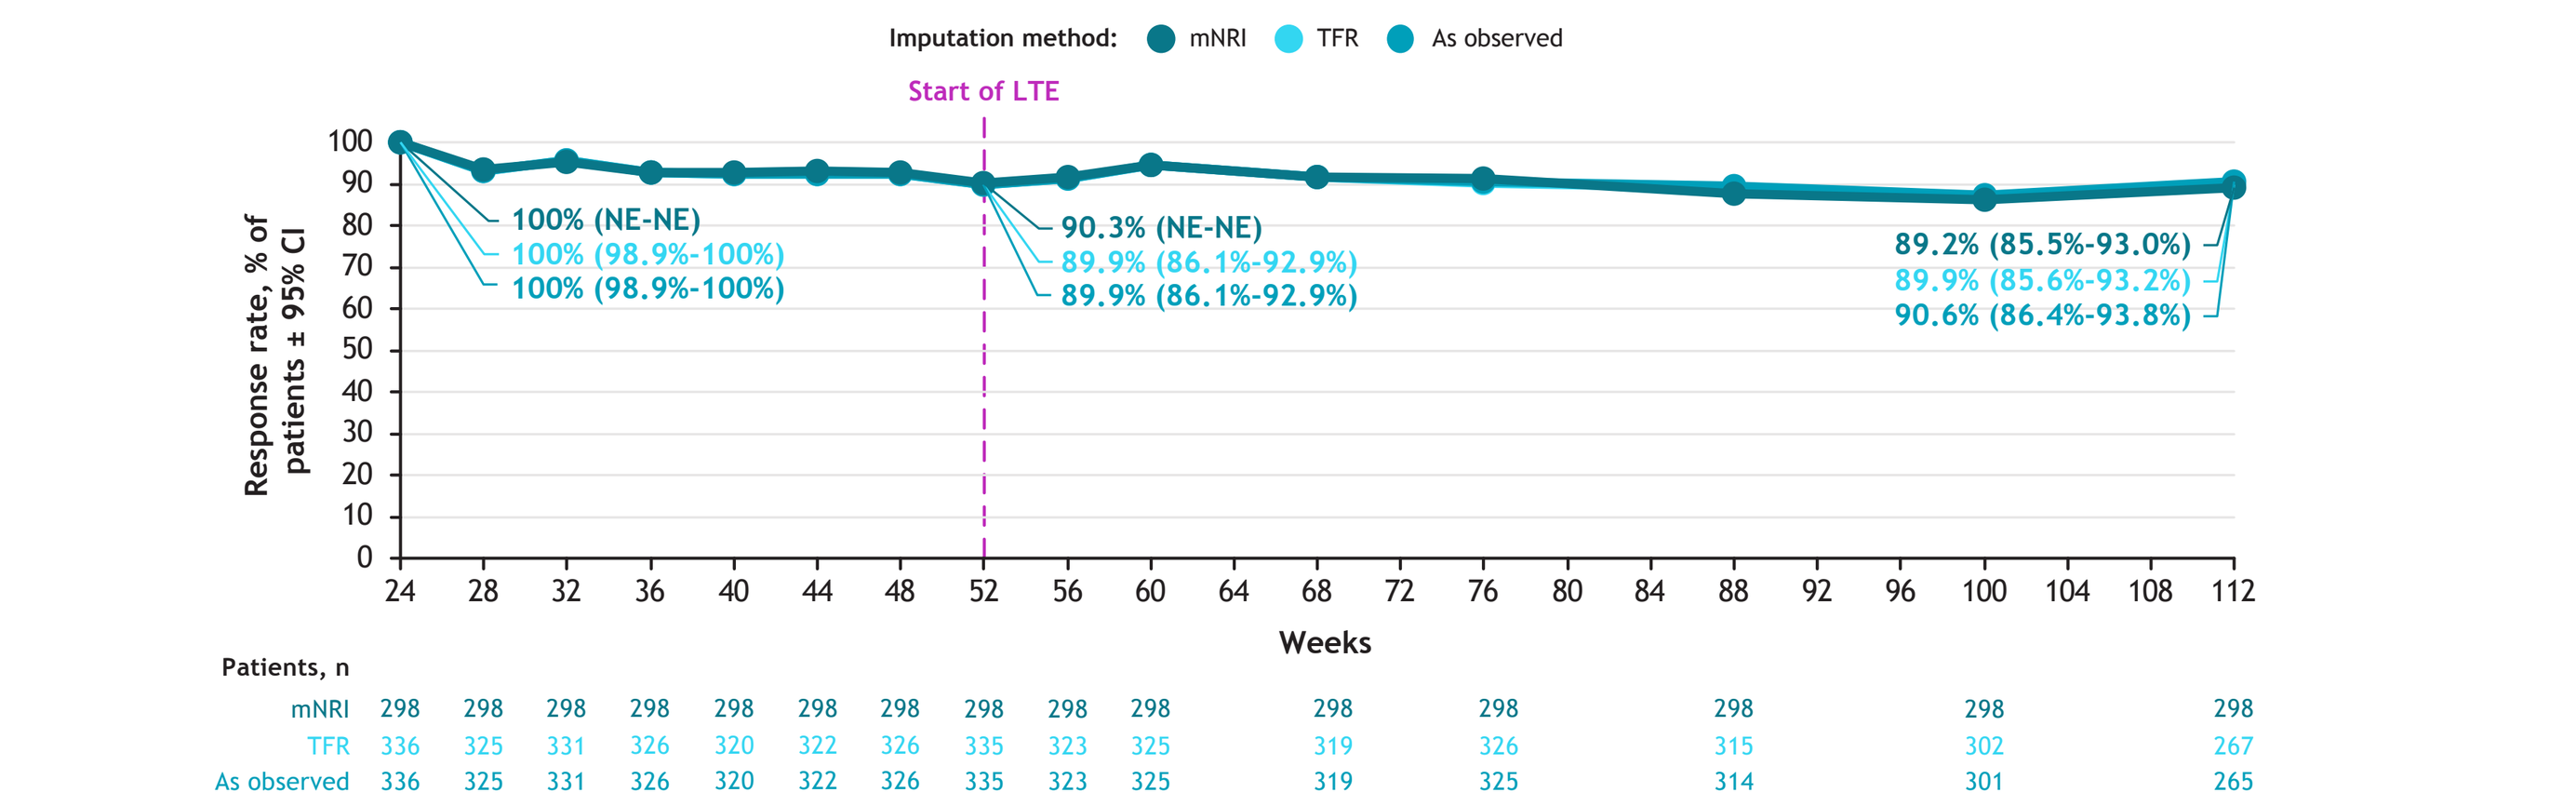

Imputation method: mNRI TFR As observed
Start of LTE
Response rate, % of patients ± 95% CI
100% (NE-NE)
100% (98.9%-100%)
100% (98.9%-100%)
90.3% (NE-NE)
89.9% (86.1%-92.9%)
89.9% (86.1%-92.9%)
89.2% (85.5%-93.0%)
89.9% (85.6%-93.2%)
90.6% (86.4%-93.8%)
Weeks
Patients, n
mNRI 298 298 298 298 298 298 298 298 298 298 298 298 298 298 298
TFR 336 325 331 326 320 322 326 335 323 325 319 326 315 302 267
As observed 336 325 331 326 320 322 326 335 323 325 319 325 314 301 265

CI, confidence interval; LTE, long-term extension; mNRI, modified nonresponder imputation; NE, not estimable; PASI 75, ≥75% reduction from baseline in Psoriasis Area and Severity Index; TFR, treatment failure rules.

Figure 7. PASI 90 response rates in Week 24 PASI 75 responders who received continuous deucravacitinib treatment from baseline until Week 112

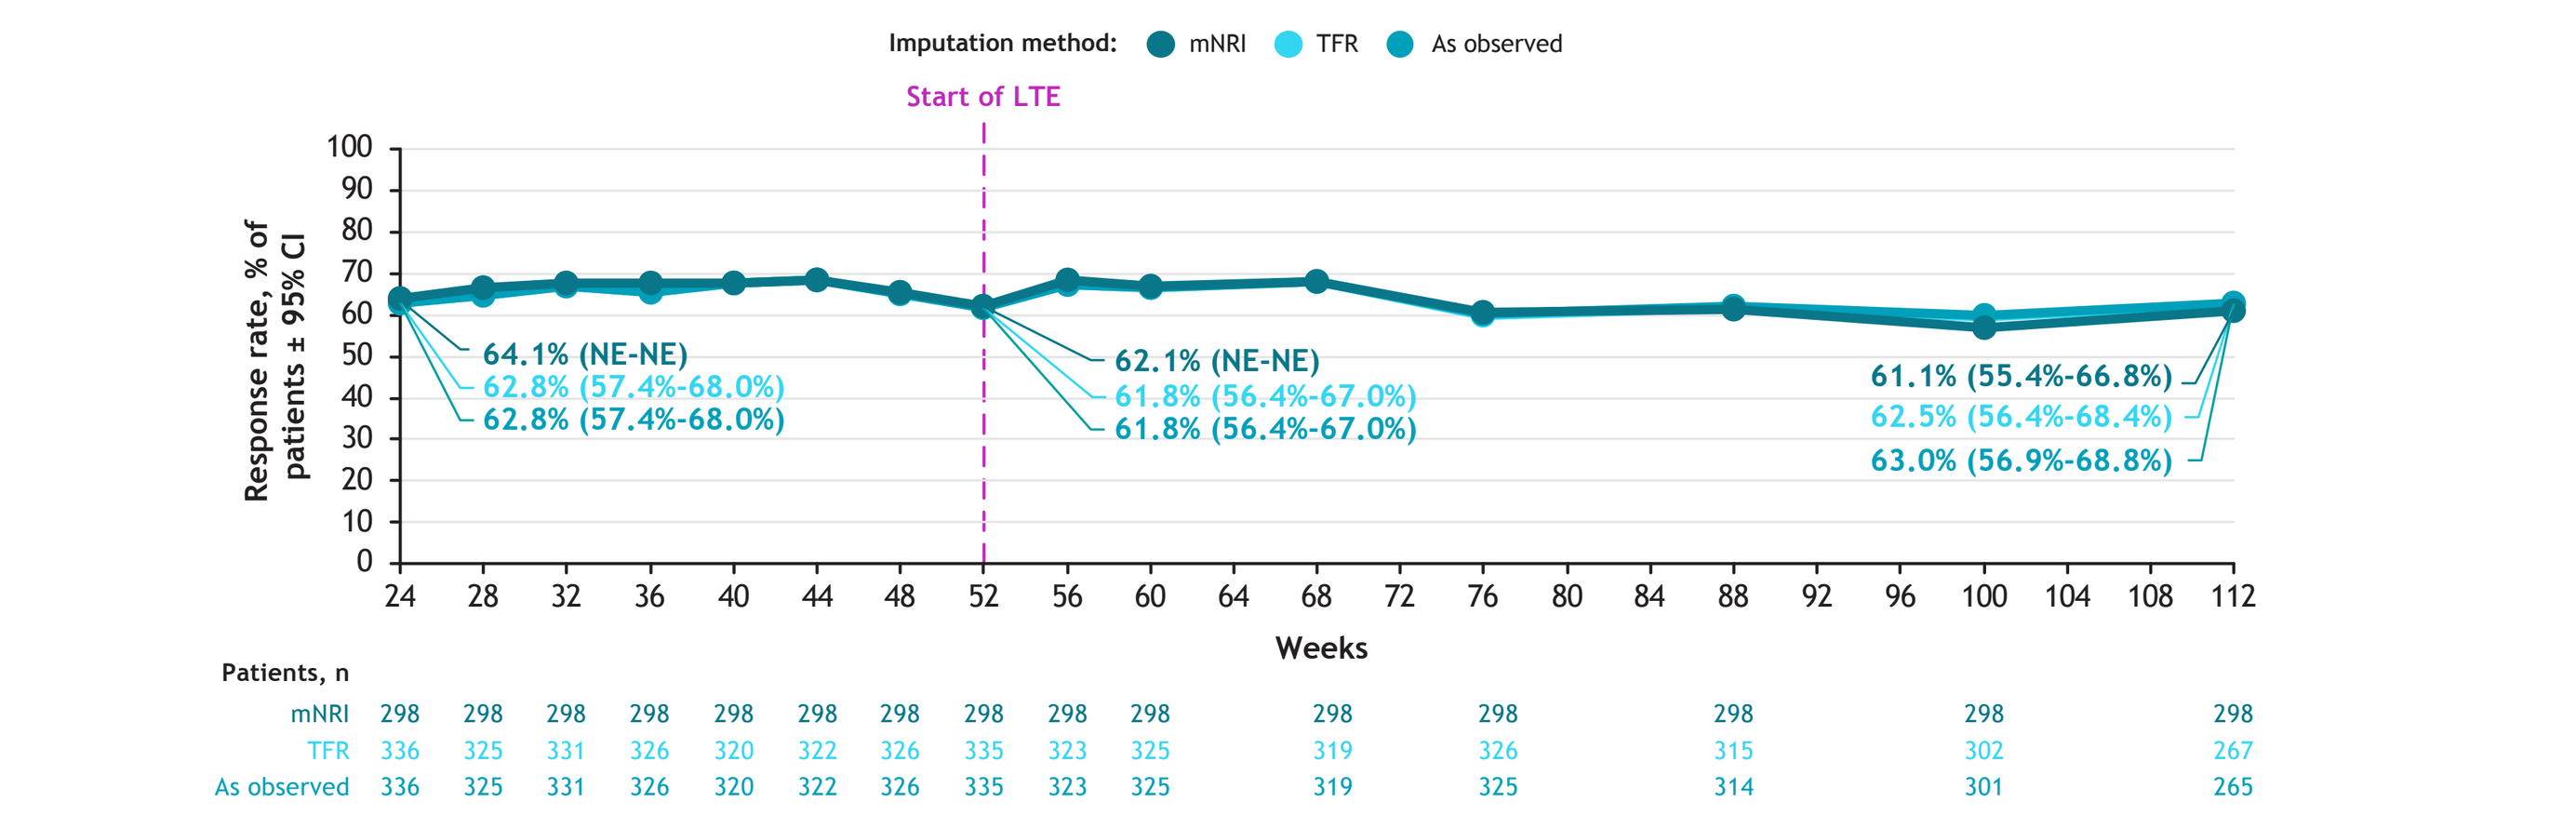

Imputation method: mNRI TFR As observed
Start of LTE
Response rate, % of patients ± 95% CI
64.1% (NE-NE)
62.8% (57.4%-68.0%)
62.8% (57.4%-68.0%)
62.1% (NE-NE)
61.8% (56.4%-67.0%)
61.8% (56.4%-67.0%)
61.1% (55.4%-66.8%)
62.5% (56.4%-68.4%)
63.0% (56.9%-68.8%)
Weeks
Patients, n
mNRI 298 298 298 298 298 298 298 298 298 298 298 298 298 298 298
TFR 336 325 331 326 320 322 326 335 323 325 319 326 315 302 267
As observed 336 325 331 326 320 322 326 335 323 325 319 325 314 301 265

CI, confidence interval; LTE, long-term extension; mNRI, modified nonresponder imputation; NE, not estimable; PASI 90, ≥90% reduction from baseline in Psoriasis Area and Severity Index; TFR, treatment failure rules.

Figure 8. sPGA 0/1 response rates in Week 24 PASI 75 responders who received continuous deucravacitinib treatment from baseline until Week 112

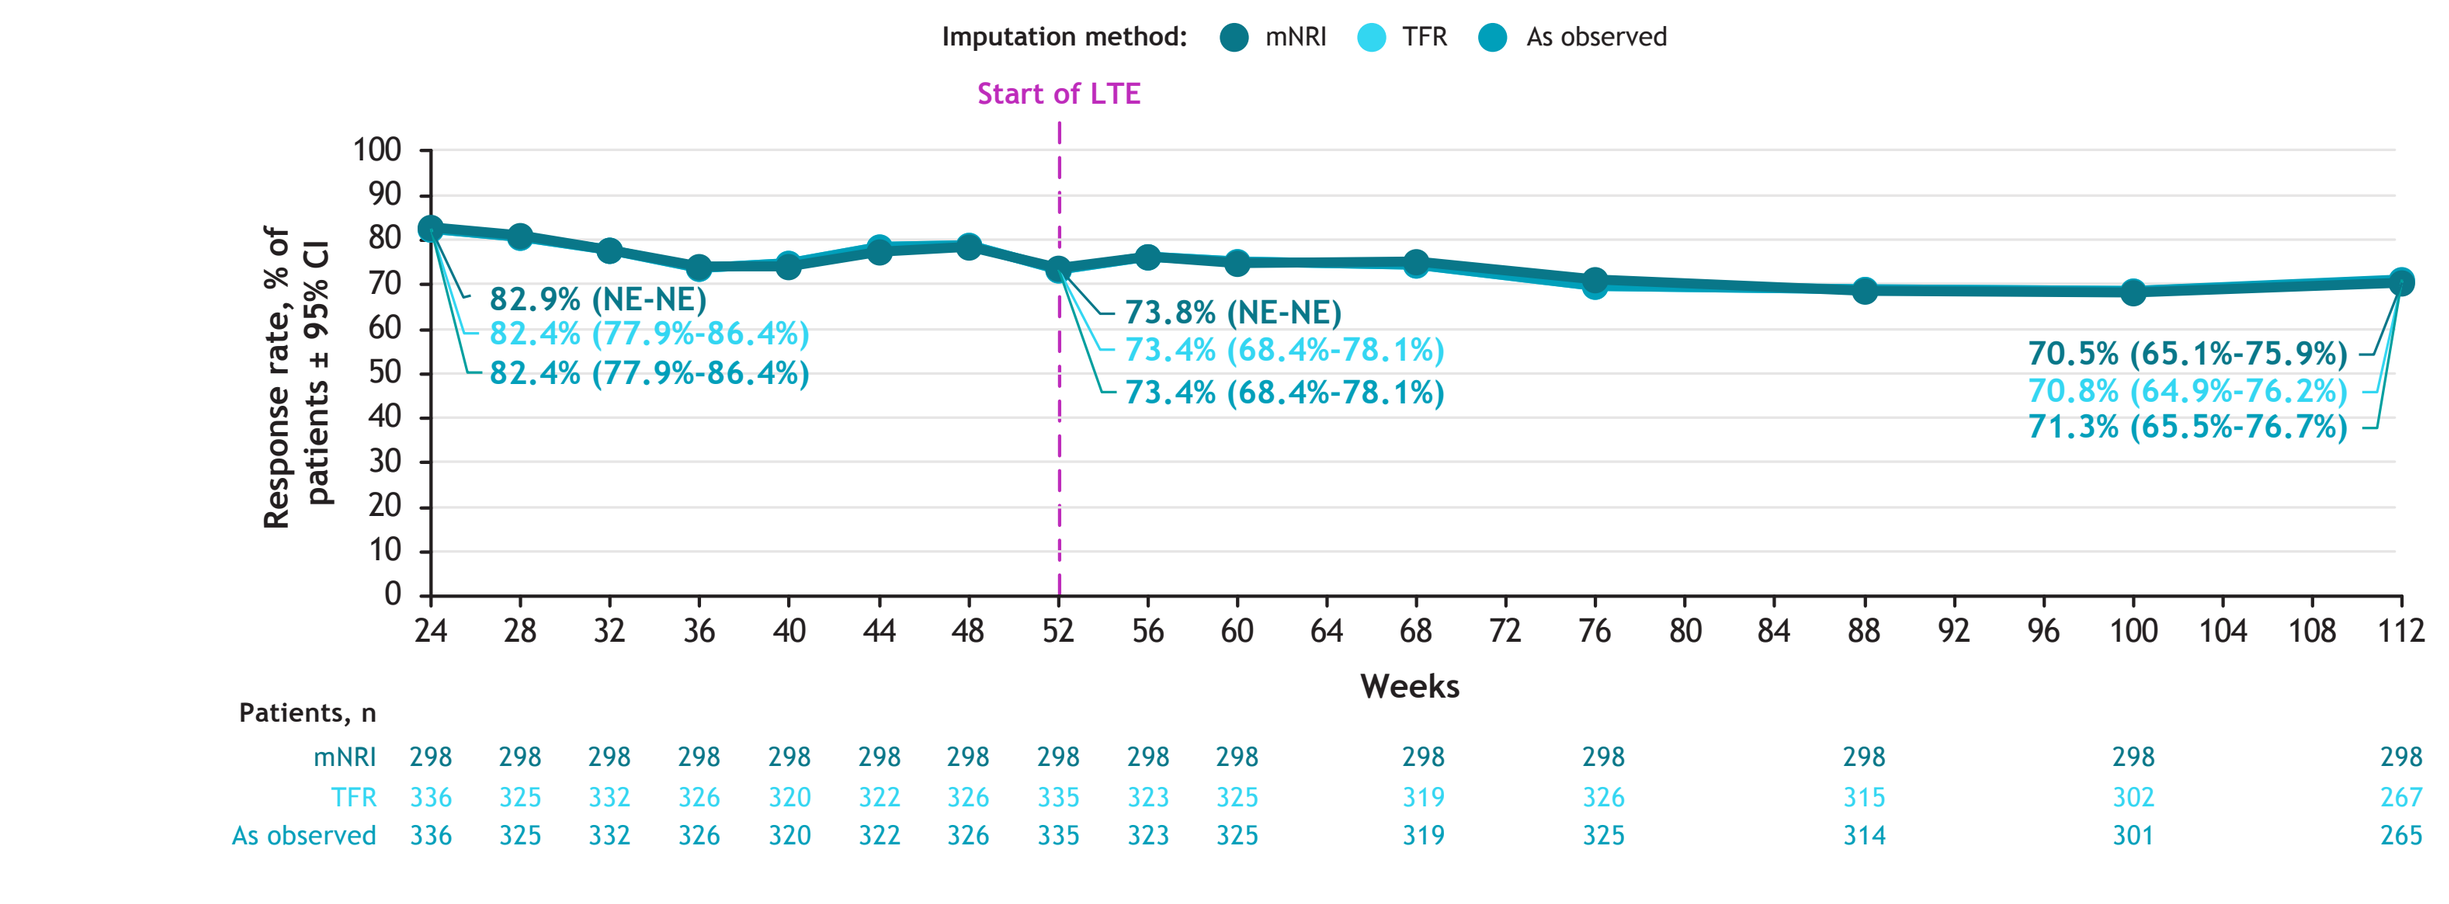

Imputation method: mNRI TFR As observed
Start of LTE
Response rate, % of patients ± 95% CI
82.9% (NE-NE)
82.4% (77.9%-86.4%)
82.4% (77.9%-86.4%)
73.8% (NE-NE)
73.4% (68.4%-78.1%)
73.4% (68.4%-78.1%)
70.5% (65.1%-75.9%)
70.8% (64.9%-76.2%)
71.3% (65.5%-76.7%)
Weeks
Patients, n
mNRI 298 298 298 298 298 298 298 298 298 298 298 298 298 298 298
TFR 336 325 332 326 320 322 326 335 323 325 319 326 315 302 267
As observed 336 325 332 326 320 322 326 335 323 325 319 325 314 301 265

CI, confidence interval; LTE, long-term extension; mNRI, modified nonresponder imputation; NE, not estimable; sPGA 0/1, static Physician Global Assessment score of 0 (clear) or 1 (almost clear) with a ≥2-point improvement from baseline; TFR, treatment failure rules.

Conclusions

- Clinical efficacy was maintained through 2 years in the pooled POETYK PSO-1/PSO-2 trials in deucravacitinib-treated patients who entered the POETYK LTE trial
 - High clinical efficacy responses (PASI 75, PASI 90, and sPGA 0/1) achieved at Week 52 were maintained overall through Week 112 in patients who received continuous deucravacitinib treatment from Day 1 and in patients who achieved PASI 75 at Week 24
 - Efficacy results were consistent across evaluated data imputation methods, including mNRI, TFR, and as-observed values
- These findings, which are consistent with previous results,[8-10] provide additional support that deucravacitinib is an efficacious long-term treatment for patients with moderate to severe plaque psoriasis

References

1. Burke JR, et al. *Sci Transl Med*. 2019;11:eaaw1736. **2.** Sotyktu [package insert]. Princeton, NJ, USA: Bristol Myers Squibb; September 2022. **3.** Sotyktu [summary of product characteristics]. Dublin, Ireland: Bristol-Myers Squibb Pharmaceutical Operations; March 2023. **4.** Sotyktu [package insert]. Tokyo, Japan: Bristol Myers Squibb K.K.; September 2022. **5.** Sotyktu [product information]. Mulgrave, VIC, Australia: Bristol Myers Squibb Australia Pty. Ltd.; December 2022. **6.** Sotyktu [product monograph]. Montreal, Canada: Bristol Myers Squibb Canada Co.; November 2022. **7.** Wrobleski ST, et al. *J Med Chem*. 2019;62:8973-8995. **8.** Armstrong AW, et al. *J Am Acad Dermatol*. 2023;88:29-39. **9.** Strober B, et al. *J Am Acad Dermatol*. 2023;88:41-50. **10.** Warren RB, et al. Presented at the 30th EADV Congress; September 29-October 2, 2021. **11.** Langley RGB, et al. *J Drugs Dermatol*. 2017;16:734-742. **12.** Papp K, et al. *Br J Dermatol*. 2021;185:1135-1145. **13.** Reich K, et al. *Br J Dermatol*. 2021;185:1146-1159.

Acknowledgments

- This study was sponsored by Bristol Myers Squibb and was supported by the NIHR Manchester Biomedical Research Centre (NIHR203308)
- Writing and editorial assistance was provided by Nick Cianciola, PhD, CMPP, of Peloton Advantage, LLC, an OPEN Health company, funded by Bristol Myers Squibb

Disclosures

DT: Research support and principal investigator (clinical trial funds to institution): AbbVie, Almirall, Amgen, Boehringer Ingelheim, Bristol Myers Squibb, Eli Lilly, Galderma, Janssen-Cilag, Leo Pharma, Novartis, Pfizer, Regeneron, Roche, Sandoz-Hexal, Sanofi, and UCB; Consultant: AbbVie, Almirall, Boehringer Ingelheim, Bristol Myers Squibb, Galapagos, Leo Pharma, Novartis, Pfizer, and UCB; Lecturer: AbbVie, Almirall, Amgen, Boehringer Ingelheim, Bristol Myers Squibb, Eli Lilly, Janssen, Leo Pharma, Novartis, Pfizer, Roche-Posay, Sanofi, Target RWE, and UCB; Scientific advisory board: AbbVie, Amgen, Boehringer Ingelheim, Bristol Myers Squibb, Eli Lilly, Janssen-Cilag, Leo Pharma, Novartis, Pfizer, Sanofi, and UCB. **ML:** Research funds on behalf of Mount Sinai: AbbVie, Amgen, Arcutis, Avotres, Boehringer Ingelheim, Cara Therapeutics, Dermavant, Eli Lilly, Incyte, Janssen, Ortho Dermatologics, Regeneron, and UCB; Consultant: Aditum Bio, Almirall, AltruBio, AnaptysBio, Arcutis, Aristea, Avotres, Boehringer Ingelheim, Brickell Biotech, Bristol Myers Squibb, Cara Therapeutics, Castle Biosciences, Celltrion, CorEvitas Psoriasis Registry, Dermavant, Dr. Reddy's Laboratories, EPI, Evommune, Facilitation of International Dermatology Education, Forte Biosciences, Foundation for Research and Education in Dermatology, Helsinn Therapeutics, Hexima, Incyte, Leo Pharma, Meiji Seika Pharma, Mindera Health, Pfizer, Seanergy, Strata, Trevi, and Verrica. **RBW:** Research grants: AbbVie, Almirall, Amgen, Celgene, Janssen, Eli Lilly, Leo Pharma, Novartis, Pfizer, and UCB; Consulting fees: AbbVie, Almirall, Amgen, Astellas, Boehringer Ingelheim, Celgene, DICE Therapeutics, Eli Lilly, GlaxoSmithKline, Janssen, Leo Pharma, Novartis, Pfizer, Sanofi, UCB, and UNION Therapeutics. **LK:** Research grants: AbbVie, Allergan, Almirall, Amgen, Arcutis, Boehringer Ingelheim, Breckinridge Pharma, Bristol Myers Squibb, Celgene, Cellceutix, Centocor, Combinatrix, Connetics, Coria, Dermavant, Dermira, Dow Pharma, Dr. Reddy's Laboratories, Eli Lilly, Galderma, Genentech, GlaxoSmithKline, Idera, Johnson & Johnson, Leo Pharma, Maruho, Merck, Medicis, Novartis AG, Pfizer, PharmaDerm, Promius, Stiefel, Sun Pharma, UCB, Valeant, and XenoPort; Honoraria: AbbVie, Allergan, Almirall, Amgen, Arcutis, Biogen Idec, Bristol Myers Squibb, Celgene, Cipher, Connetics, Dermavant, Dermira, Dr. Reddy's Laboratories, Eli Lilly, Galderma, Genentech, GlaxoSmithKline, Johnson & Johnson, Leo Pharma, Merck, Novartis AG, PharmaDerm, Promius, Serono (Merck Serono International SA), Stiefel, Sun Pharma, Taro, UCB, and Valeant. **JZ, SB, AN, RMK, and ML:** Employees and shareholders: Bristol Myers Squibb. **KH:** Consultant: Bristol Myers Squibb via Syneos Health. **AB:** Speaker (with honoraria): AbbVie, Arcutis, Bristol Myers Squibb, Eli Lilly, Pfizer, Regeneron, Sanofi, and UCB; Scientific adviser (with honoraria): AbbVie, Abcentra, Aclaris, Affibody, Aligos, Almirall, Alumis, Amgen, AnaptysBio, Apogee, Arcutis, Arena, Aslan, Athenex, Bluefin Biomedicine, Boehringer Ingelheim, Bristol Myers Squibb, Cara Therapeutics, CTI BioPharma, Dermavant, EcoR1, Eli Lilly, Escient, Evelo Biosciences, Evommune, Forte Biosciences, Galderma, HighlightII Pharma, Incyte, InnoventBio, Janssen, Landos, Leo Pharma, Lipidio, Merck, Nektar, Novartis, Pfizer, Rani, Rapt, Regeneron, Sanofi Genzyme, Spherix Global Insights, Sun Pharma, Takeda, TLL Pharmaceutical, TrialSpark, UCB, UNION Therapeutics, Ventyx Biosciences, Vibliome, and Xencor; Clinical study investigator (clinical study funds received by institution): AbbVie, Acelyrin, Allakos, Almirall, Alumis, Amgen, Arcutis, Athenex, Boehringer Ingelheim, Bristol Myers Squibb, Concert, Dermavant, Eli Lilly, Evelo Biosciences, Evommune, Galderma, Incyte, Janssen, Leo Pharma, Merck, Novartis, Pfizer, Regeneron, Sanofi, Sun Pharma, UCB, and Ventyx Biosciences.

Presented at the 2nd Annual Dermatology Innovation Symposium™; September 8-10, 2023; Anaheim, California
This is an encore of the 2023 European Academy of Dermatology and Venereology Spring poster

Email: diamant.thaci@uksh.de

Copies of this poster are for personal use only and may not be reproduced without written permission of the authors.